P4-07-20

Distance to Treating Oncologist as Potential Prognostic Real-World-Factor for Patients with HR+/HER2– Advanced Breast Cancer – Results from the Non-Interventional Study PERFORM

Michael P. Lux[1], Georg Pfeiler[2], Matthias Korell[3], Julia Radosa[4], Thomas Decker[5], Mustafa Deryal[6], Thomas Fietz[7], Andreas Köhler[8], Björn Schöttker[9], Jochen Wilke[10], Jan Knoblich[11], Volker Petersen[12], Natalija Deuerling[13], Thomas Gabrysiak[14], Uwe Rhein[15], Michael Maasberg[16], Thomas Resch[17], Denise Wrobel[18], Reinhard Depenbusch[19], Isolde Gröll[20], Vesna Bjelic-Radisic[21], Thomas Göhler[22], Nikola Bangemann[23], Daniel Egle[24], Lutz Jacobasch[25], Lothar Müller[26], Gabriele Prange-Krex[27], Oliver Tomé[28], Cristoph Großmann[29], Petra Krabisch[30], Henriette Princk[31], Johanna Dzieran[32], Katja Gratzke[33], Martin Glasstetter[34], Anne Adams[32], Esther Glastetter[32], André Breitbach[32], Rupert Bartsch[34]

[1]Department for Gynecology and Obstetrics, Frauenklinik St. Louise, Paderborn, Frauenklinik St. Josefs-Krankenhaus, Salzkotten, St. Vincenz-Hospital GmbH, Paderborn, Germany; [2]Department of Obstetrics and Gynecology, Division of General Gynecology and Gynecologic Oncology, Medical University of Vienna, Vienna, Austria; [3]Department for Gynecology, Johanna Etienne Krankenhaus Neuss, Neuss, Germany; [4]Department for Obstetrics, Universitätsklinikum des Saarlandes, Homburg (Saar), Germany; [5]Practice for Hematology and Internistic Oncology, Ravensburg, Germany; [6]Caritasklinikum Frauenklinik, Saarbrücken, Germany; [7]Practice for Hematology and Internistic Oncology, Gastroenterology, Singen, Germany; [8]Hematologic-Oncologic Practice Langen, Langen, Germany; [9]Hematologic-Oncologic Practice Würzburg, Würzburg, Germany; [10]Practice for Hematology and Internistic Oncology, Fürth, Germany; [11]Oncology Practice, Lörrach, Germany; [12]Practice for Hematology and Internistic Oncology, Heidenheim a. d. B.; Germany; [13]Department for Gynecology and Obstetrics, Frauenklinik, Klinikum Fichtelgebirge, Marktredwitz, Germany; [14]Hematologic-Oncologic Practice Wolfsburg, Wolfsburg, Germany; [15]Department for Gynecology and Obstetrics, SRH Zentralklinikum Suhl, Suhl, Germany; [16]Practice for Internal Medicine, Hematology/Internistic Oncology, Palliative Medicine and Geriatrics, Mayen, Germany; [17]Practice for Gynecology and Obstetrics, Brandenburg a. d. Havel, Germany; [18]Practice for Gynecology and Obstetrics, Bamberg, Germany; [19]Practice for Internal Medicine, Hematology/Internistic Oncology and Palliative Medicine; [20]Practice for Gynecology and Obstetrics, Ebersberg; [21]Department Senology Helios University Hospital Wuppertal, University Witten/Herdecke, Wuppertal, Germany; [22]Practice for Internal Medicine, Hematology/Internistic Oncology and Palliative Medicine, Dresden/Freiburg/Meißen, Germany; [23]Department for Gynecology and Obstetrics, Medical University Lausitz, Carl Thiem, Cottbus, Germany; [24]Department for Gynecology and Obstetrics, Universitätsklinik für Gynäkologie und Geburtshilfe, Innsbruck, Austria; [25]Practice for Internal Medicine, Hematology, Oncology and Gastroenterology,Desden, Germany; [26]Practice for Internal Medicine, Hematology, Oncology, Palliative Medicine and Rescue Medicine, Leer, Germany; [27]Practice for Internal Medicine, Hematology, Oncology and Palliative Medicine, Dresden, Germany; [28]Practice for Gynecology and Obstetrics, Karlsruhe, Germany; [29]Practice for Gynecology and Obstetrics, Hamburg, Germany; [30]Department for Gynecology and Obstetrics, Klinikum Chemnitz, Chemnitz, Germany; [31]Department for Gynecology and Obstetrics, Universitätsklinikum Schleswig-Holstein, Lübeck, Germany; [32]Pfizer Pharma GmbH, Berlin, Germany; [33]iOMEDICO, Freiburg, Germany; [34]Department of Medicine I, Division of Oncology, Medical University of Vienna, Vienna, Austria

Objective

To investigate whether distance to treating oncologist (DTO) influence toxicity, therapy-management and disease progression of patients with HR+/HER2– advanced breast cancer (ABC) in the PERFORM study.

Conclusions

Our results generally support the use of palbociclib plus endocrine therapy (ET) as a relevant first-line (1L) therapy option regardless of age and DTO in patients with HR+/HER2– ABC. Moreover, our results indicate that the DTO (travel burden) might be a relevant real-world factor influencing outcome,that warrants further analyses including patient-reported outcomes with longer follow-up as well as external validation.

Background

ET in combination with cyclin-dependent-kinase 4/6 inhibitors is the 1L standard of care for patients with HR+/HER2– advanced or metastatic breast cancer (ABC). Efficacy, safety and tolerability have been demonstrated in pivotal phase III randomized clinical trials[1–5], typically carried out under strictly defined conditions. Real-world analyses are intended to contribute to the consideration of the manageability and effectiveness of approved therapy concepts in existing care structures in broader real-world populations.[6] Factors that can influence the safety, tolerability, management of a therapy and thus effectiveness and quality of life in the reality of care can also be considered or identified. A potential real-world factor that is rarely considered is the distance of the patient's residence to the treating oncologist. Therefore, an exploratory analysis was carried out as part of the third interim analysis (IA3) of the non-interventional study (NIS) PERFORM, to consider a possible influence of this parameter on toxicity, therapy-management and disease progression.

Methods

The prospective NIS PERFORM is carried out in approx. 240 urban and rural study sites across Germany and Austria. More than 1400 patients with HR+/HER2– ABC treated with palbociclib plus ET in the 1L setting are currently enrolled. Key eligibility criteria are: diagnosis of HR+/HER2– ABC, 1L treatment with palbociclib + ET as per local product label, age of 18 years or older, and no prior treatment for advanced disease.[7] The primary endpoint is 1L progression-free survival (PFS), defined as start of 1L treatment to first progression or death, whichever comes first. Patients without tumor progression or death at the time of analysis are censored at their last date of last contact or at the start date of a second-line (2L) therapy, whichever comes first. Secondary endpoints include treatment patterns, effectiveness (including outcomes in 2L and third-line [3L] treatment), treatment expectation/satisfaction, potential impact of socioeconomic status on outcomes and assessment of quality of life as well as patterns of biomarker analyses and genetic testing.[7]

Three years after first patient enrollment, IA3 was conducted with data cutoff in September 2023. Demographic and disease characteristics as well as socioeconomic information, including DTO, are documented at baseline. Adverse events (AEs) and therapy modifications are continuously documented. Disease progression is evaluated according to routine clinical practice. Here, we focus on subgroups with DTO < 20 km and ≥ 20 km with additional age-stratification (< 75 and ≥ 75 years). Patient-, disease- and socioeconomic characteristics, AEs, therapy modifications and real-world PFS (rwPFS) rates at 6, 12, 18 and 24 months are included in this analysis. Descriptive statistics are used to summarize results. PFS-analyses are based on Kaplan–Meier estimation. Multivariable or other types of analyses controlling for potential confounders have not been done.

Results

PATIENT CHARACTERISTICS & SOCIOECONOMIC FACTORS

In IA3 of the NIS PERFORM, 990 patients are evaluable for analysis. Characteristics of the overall population were previously reported.[8-10] Of these, 854 patients are evaluable for subgroup analysis regarding DTO. In both DTO subgroups, approx. 92% of patients are postmenopausal, 7% are pre-/perimenopausal and 1% are men. There are considerably more patients living in a distance < 20 km to treating oncologist (612 vs. 242). Those patients with a DTO < 20 km show a tendency to be older compared with those with higher DTO (178 of 612 [29.1%] vs. 60 of 242 [24.8%] are ≥ 75 years). Among patients < 75 years, approx. 22% are full-time employed and approx. 17% are part-time employed, irrespective of DTO. The number of children is comparable among age groups, irrespective of DTO. However, there is a trend that patients with a DTO < 20 km are more likely to live alone compared to patients with a DTO ≥ 20 km, which is particularly true for those who are 75 years or older (57.3% vs. 41.7%) (**Table 1**).

TUMOR CHARACTERISTICS

Tumor stages and number of metastatic sites seem largely comparable among the subgroups, irrespective of age. Patients with a DTO ≥ 20 km appear to be slightly more likely to present with *de novo* ABC compared to patients with a DTO < 20 km (42.1% vs. 36.4%). This tendency is mainly driven by patients aged 75 years or older (58.3% vs. 39.3%). Moreover, there is a tendency that patients under 75 years are more likely to have visceral metastasis in the DTO < 20 km subgroup compared to those with a DTO of ≥ 20 km (48.2% vs. 42.9%) (**Table 2**).

Table 1. Patient and socioeconomic characteristics at inclusion by distance to treating oncologist and age

	DTO < 20 km			DTO ≥ 20 km		
	Total (n=612)	**< 75 years (n=434)**	**≥ 75 years (n=178)**	**Total (n=242)**	**< 75 years (n=182)**	**≥ 75 years (n=60)**
Age at start of 1L treatment, years						
Median (Q1-Q3)	68.60 (59.04-76.83)	63.00 (55.78-69.34)	79.41 (77.56-81.97)	66.53 (58.18-74.70)	62.06 (55.81-68.09)	79.50 (77.74-82.32)
Sex, n (%)						
Female	607 (99.2)	431 (99.3)	176 (98.9)	240 (99.2)	181 (99.5)	59 (98.3)
Male	5 (0.8)	3 (0.7)	2 (1.1)	2 (0.8)	1 (0.5)	1 (1.7)
Menopausal status, n (%)						
Pre-/perimenopausal	45 (7.3)	45 (10.3)	0 (0.0)	19 (7.8)	19 (10.4)	0 (0.0)
Postmenopausal	562 (91.8)	386 (88.9)	176 (98.9)	221 (91.3)	162 (89.0)	59 (98.3)
ECOG performance status, n (%)						
0	285 (46.6)	228 (52.5)	57 (32.0)	122 (50.4)	98 (53.8)	24 (40.0)
1	249 (40.7)	167 (38.5)	82 (46.1)	90 (37.2)	61 (33.5)	29 (48.3)
≥ 2	61 (10.0)	29 (6.7)	32 (18.0)	26 (10.7)	19 (10.4)	7 (11.7)
No assessment done/missing	17 (2.8)	10 (2.3)	7 (4.0)	4 (1.7)	4 (2.2)	0 (0.0)
Occupation, n (%)						
Employed full-time	94 (15.4)	93 (21.4)	1 (0.6)	41 (16.9)	41 (22.5)	0 (0.0)
Employed part-time[a]	74 (12.1)	72 (16.6)	2 (1.1)	31 (12.8)	30 (16.5)	1 (1.7)
Not gainfully employed/retired	383 (62.6)	227 (52.3)	156 (87.6)	152 (62.8)	99 (54.4)	53 (88.3)
Missing/not derivable	61 (10.0)	42 (9.7)	19 (10.7)	18 (7.4)	12 (6.6)	6 (10.0)
No. of additional persons in household, n (%)						
0	250 (40.8)	148 (34.1)	102 (57.3)	72 (29.8)	47 (25.8)	25 (41.7)
≤ 3 other persons	314 (51.3)	248 (57.1)	66 (37.1)	147 (60.7)	116 (63.7)	31 (51.7)
> 3 other persons	17 (2.8)	15 (3.5)	2 (1.1)	7 (2.9)	7 (3.8)	0 (0.0)
Missing/not derivable	31 (5.1)	23 (5.3)	8 (4.5)	16 (6.6)	12 (6.6)	4 (6.7)
No. of children, n (%)						
0	92 (15.0)	79 (18.2)	13 (7.3)	35 (14.5)	30 (16.5)	5 (8.3)
1 or 2	383 (62.6)	272 (62.7)	111 (62.4)	145 (59.9)	110 (60.4)	35 (58.3)
≥ 3	101 (16.5)	62 (14.3)	39 (21.9)	50 (20.7)	32 (17.6)	18 (30.0)
Missing	36 (5.9)	21 (4.8)	15 (8.4)	12 (5.0)	10 (5.5)	2 (3.3)

[a]including primary / secondary occupation.
1L, first-line; DTO, distance to treating oncologist; Q, quartile.

Table 2. Tumor characteristics at inclusion by distance to treating oncologist and age

	DTO < 20 km			DTO ≥ 20 km		
	Total (n=612)	**< 75 years (n=434)**	**≥ 75 years (n=178)**	**Total (n=242)**	**< 75 years (n=182)**	**≥ 75 years (n=60)**
Time since initial diagnosis, years						
Median (Q1-Q3)	3.46 (0.13-10.89)	3.28 (0.13-10.07)	4.00 (0.11-13.05)	2.36 (0.10-9.18)	2.97 (0.13-8.78)	0.16 (0.09-9.68)
Tumor stage, n (%)						
Locoregionally advanced	31 (5.1)	15 (3.5)	16 (9.0)	16 (6.6)	11 (6.0)	5 (8.3)
Metastatic	580 (94.8)	418 (96.3)	173 (91.0)	225 (93.0)	171 (94.0)	54 (90.0)
Missing	1 (0.2)	1 (0.2)	0 (0.0)	1 (0.4)	0 (0.0)	1 (1.7)
De novo advanced breast cancer, n (%)						
Yes	223 (36.4)	153 (35.3)	70 (39.3)	102 (42.1)	67 (36.8)	35 (58.3)
No	389 (63.6)	281 (64.7)	108 (60.7)	140 (57.9)	115 (63.2)	25 (41.7)
No. of metastatic sites present, n (%)						
0[a]	46 (7.5)	27 (6.2)	19 (10.7)	26 (10.7)	18 (9.9)	8 (13.3)
1	378 (61.8)	269 (62.0)	109 (61.2)	139 (57.4)	104 (57.1)	35 (58.3)
2	114 (18.6)	78 (18.0)	36 (20.2)	54 (22.3)	42 (23.1)	12 (20.0)
3	57 (9.3)	46 (10.6)	11 (6.2)	15 (6.2)	12 (6.6)	3 (5.0)
≥ 4	17 (2.8)	14 (3.2)	3 (1.7)	8 (3.3)	6 (3.3)	2 (3.3)
Disease site, n (%)						
Visceral[b]	286 (46.7)	209 (48.2)	77 (43.3)	106 (43.8)	78 (42.9)	28 (46.7)
Non-visceral only[c] (excl. bone only)	69 (11.3)	41 (9.4)	28 (15.7)	29 (12.0)	23 (12.6)	6 (10.0)
Bone only	211 (34.5)	157 (36.2)	54 (30.3)	81 (33.5)	63 (34.6)	18 (30.0)
No metastases present at inclusion[a]	46 (7.5)	27 (6.2)	19 (10.7)	26 (10.7)	18 (9.9)	8 (13.3)

[a]patients with locoregionally advanced disease or metastases, that were removed before inclusion (e.g. radiation, surgery)
[b]visceral sites: all metastatic sites excluding non-visceral sites and bone only (e.g. lung, liver, pleura, peritoneum, brain)
[c]non-visceral sites (excl. bone only): lymph-nodes (distant, regional), skin, soft tissue.
DTO, distance to treating oncologist; Q, quartile.

ADVERSE EVENTS AND THERAPY MODIFICATIONS

A comparable relative frequency of AEs was observed for PERFORM patients, regardless of DTO. Palbociclib-related AEs appeared to be slightly more common among patients of 75 years or older. Serious AEs were also documented more frequently for older patients irrespective of DTO. Palbociclib-related serious AEs were generally low in both subgroups, regardless of age (2.2%–5.0%). Therapy modifications for palbociclib treatment occurred with a comparable frequency among subgroups with a DTO of < 20 km or ≥ 20 km, respectively (73.5% vs. 71.5%). In both subgroups, approx. 10% of patients discontinued palbociclib based therapy due to AEs that emerged during treatment (TEAEs). 3.9% and 2.9% of patients with a DTO of < 20 km or ≥ 20 km discontinued therapy due to palbociclib related AEs (**Table 3**).

PROGRESSION-FREE SURVIVAL

34.0% and 38.0% of patients experienced disease progression or death in the subgroups DTO < 20 km and DTO ≥ 20 km, respectively. 57.5% of patients in subgroup DTO < 20 km and 52.5% of patients in subgroup DTO ≥ 20 km are still undergoing treatment. Generally, PFS rates seem comparable among both groups, irrespective of age. However, there could be a trend towards slightly lower PFS-rates in patients with a DTO of ≥ 20 km compared to patients with a DTO of < 20 km, regardless of age. The lowest 24-month PFS-rate of 32.5% was observed in older patients with DTO ≥ 20 km, while the overall 24-month PFS rate in the DTO subgroup < 20 km and ≥ 20 km amounted to 55.1% and 46.7%, respectively; this observation might be especially influenced by low sample size, limited follow-up time and potential confounders and therefore requires further investigation (**Table 4, Figure 1**).

Table 3. Adverse events and therapy modifications

	DTO < 20 km			DTO ≥ 20 km		
	Total (n=612)	**< 75 years (n=434)**	**≥ 75 years (n=178)**	**Total (n=242)**	**< 75 years (n=182)**	**≥ 75 years (n=60)**
AEs, n (%)						
TEAE	519 (84.8)	361 (83.2)	158 (88.8)	209 (86.4)	158 (86.8)	51 (85)
Serious TEAE	168 (27.5)	112 (25.8)	56 (31.5)	65 (26.9)	46 (25.3)	19 (31.7)
Grade 1/2 TEAE	456 (74.5)	316 (72.8)	140 (78.7)	182 (75.2)	138 (75.8)	44 (73.3)
Grade 3/4 TEAE	303 (49.5)	206 (47.5)	97 (54.5)	121 (50)	86 (47.3)	35 (58.3)
TEAE leading to discontinuation of palbociclib	59 (9.6)	41 (9.4)	18 (10.1)	25 (10.3)	17 (9.3)	8 (13.3)
Palbociclib-related AE	386 (63.1)	259 (59.7)	127 (71.3)	157 (64.9)	115 (63.2)	42 (70)
Palbociclib-related SAE	20 (3.3)	13 (3.0)	7 (3.9)	7 (2.9)	4 (2.2)	3 (5.0)
Palbociclib-related grade 3/4 AE	200 (32.7)	143 (32.9)	57 (32)	77 (31.8)	52 (28.6)	25 (41.7)
Palbociclib-related AE leading to discontinuation of palbociclib	24 (3.9)	17 (3.9)	7 (3.9)	7 (2.9)	2 (1.1)	5 (8.3)
Therapy modifications palbociclib[a], n (%)						
Yes	450 (73.5)	312 (71.9)	138 (77.5)	173 (71.5)	127 (69.8)	46 (76.7)
No	162 (26.5)	122 (28.1)	40 (22.5)	69 (28.5)	55 (30.2)	14 (23.3)
Yes - Dose modified	243 (39.7)	165 (38.0)	78 (43.8)	87 (36.0)	57 (31.3)	30 (50.0)

[a]including dose modifications, interruptions within / between cycles or skipped cycles.
AE, adverse event; TEAE, treatment-emergent adverse event; SAE, serious adverse event.

Table 4. PFS-rates and reasons for end of 1L treatment

	DTO < 20 km			DTO ≥ 20 km		
	Total (n=612)	**< 75 years (n=434)**	**≥ 75 years (n=178)**	**Total (n=242)**	**< 75 years (n=182)**	**≥ 75 years (n=60)**
Reasons for end of 1L treatment, n (%)						
Progressive disease	172 (28.1)	133 (30.6)	39 (21.9)	75 (31.0)	61 (33.5)	14 (23.3)
Serious adverse event	36 (5.9)	21 (4.8)	15 (8.4)	14 (5.8)	8 (4.4)	6 (10.0)
Lost to follow-up	21 (3.4)	16 (3.7)	5 (2.8)	9 (3.7)	6 (3.3)	3 (5.0)
Withdrawal of informed consent	8 (1.3)	5 (1.2)	3 (1.7)	7 (2.9)	7 (3.8)	0 (0.0)
Other	23 (3.8)	17 (3.9)	6 (3.4)	10 (4.1)	7 (3.8)	3 (5.0)
Still under treatment	352 (57.5)	242 (55.8)	110 (61.8)	127 (52.5)	93 (51.1)	34 (56.7)
1L Progression-free survival						
Events, n (%)	208 (34.0)	154 (35.5)	54 (30.3)	92 (38.0)	74 (40.7)	18 (30.0)
6-month rate in % (95% CI)	85.9 (82.8, 88.4)	85.5 (81.8, 88.6)	86.7 (80.6, 90.9)	82.7 (77.2, 86.9)	81.9 (75.4, 86.9)	84.8 (72.8, 91.8)
12-month rate in % (95% CI)	73.2 (69.2, 76.7)	72.3 (67.6, 76.5)	75.2 (67.7, 81.2)	71.0 (64.4, 76.6)	69.9 (62.2, 76.3)	74.3 (60.2, 84.0)
18-month rate in % (95% CI)	64.2 (59.8, 68.3)	61.9 (56.5, 66.8)	70.1 (61.9, 76.8)	59.8 (52.2, 66.6)	58.3 (49.5, 66.1)	65.1 (48.7, 77.4)
24-month rate in % (95% CI)	55.1 (49.7, 60.1)	52.5 (45.9, 58.7)	61.5 (52.0, 69.7)	46.7 (37.1, 55.7)	45.7 (35.6, 55.2)	32.5 (2.0, 72.5)

1L, first-line; CI, confidence interval; DTO, distance to treating oncologist; PFS, progression-free survival.

Figure 1. Progression-free survival rates of age-stratified subgroups DTO < 20 km and ≥ 20 km

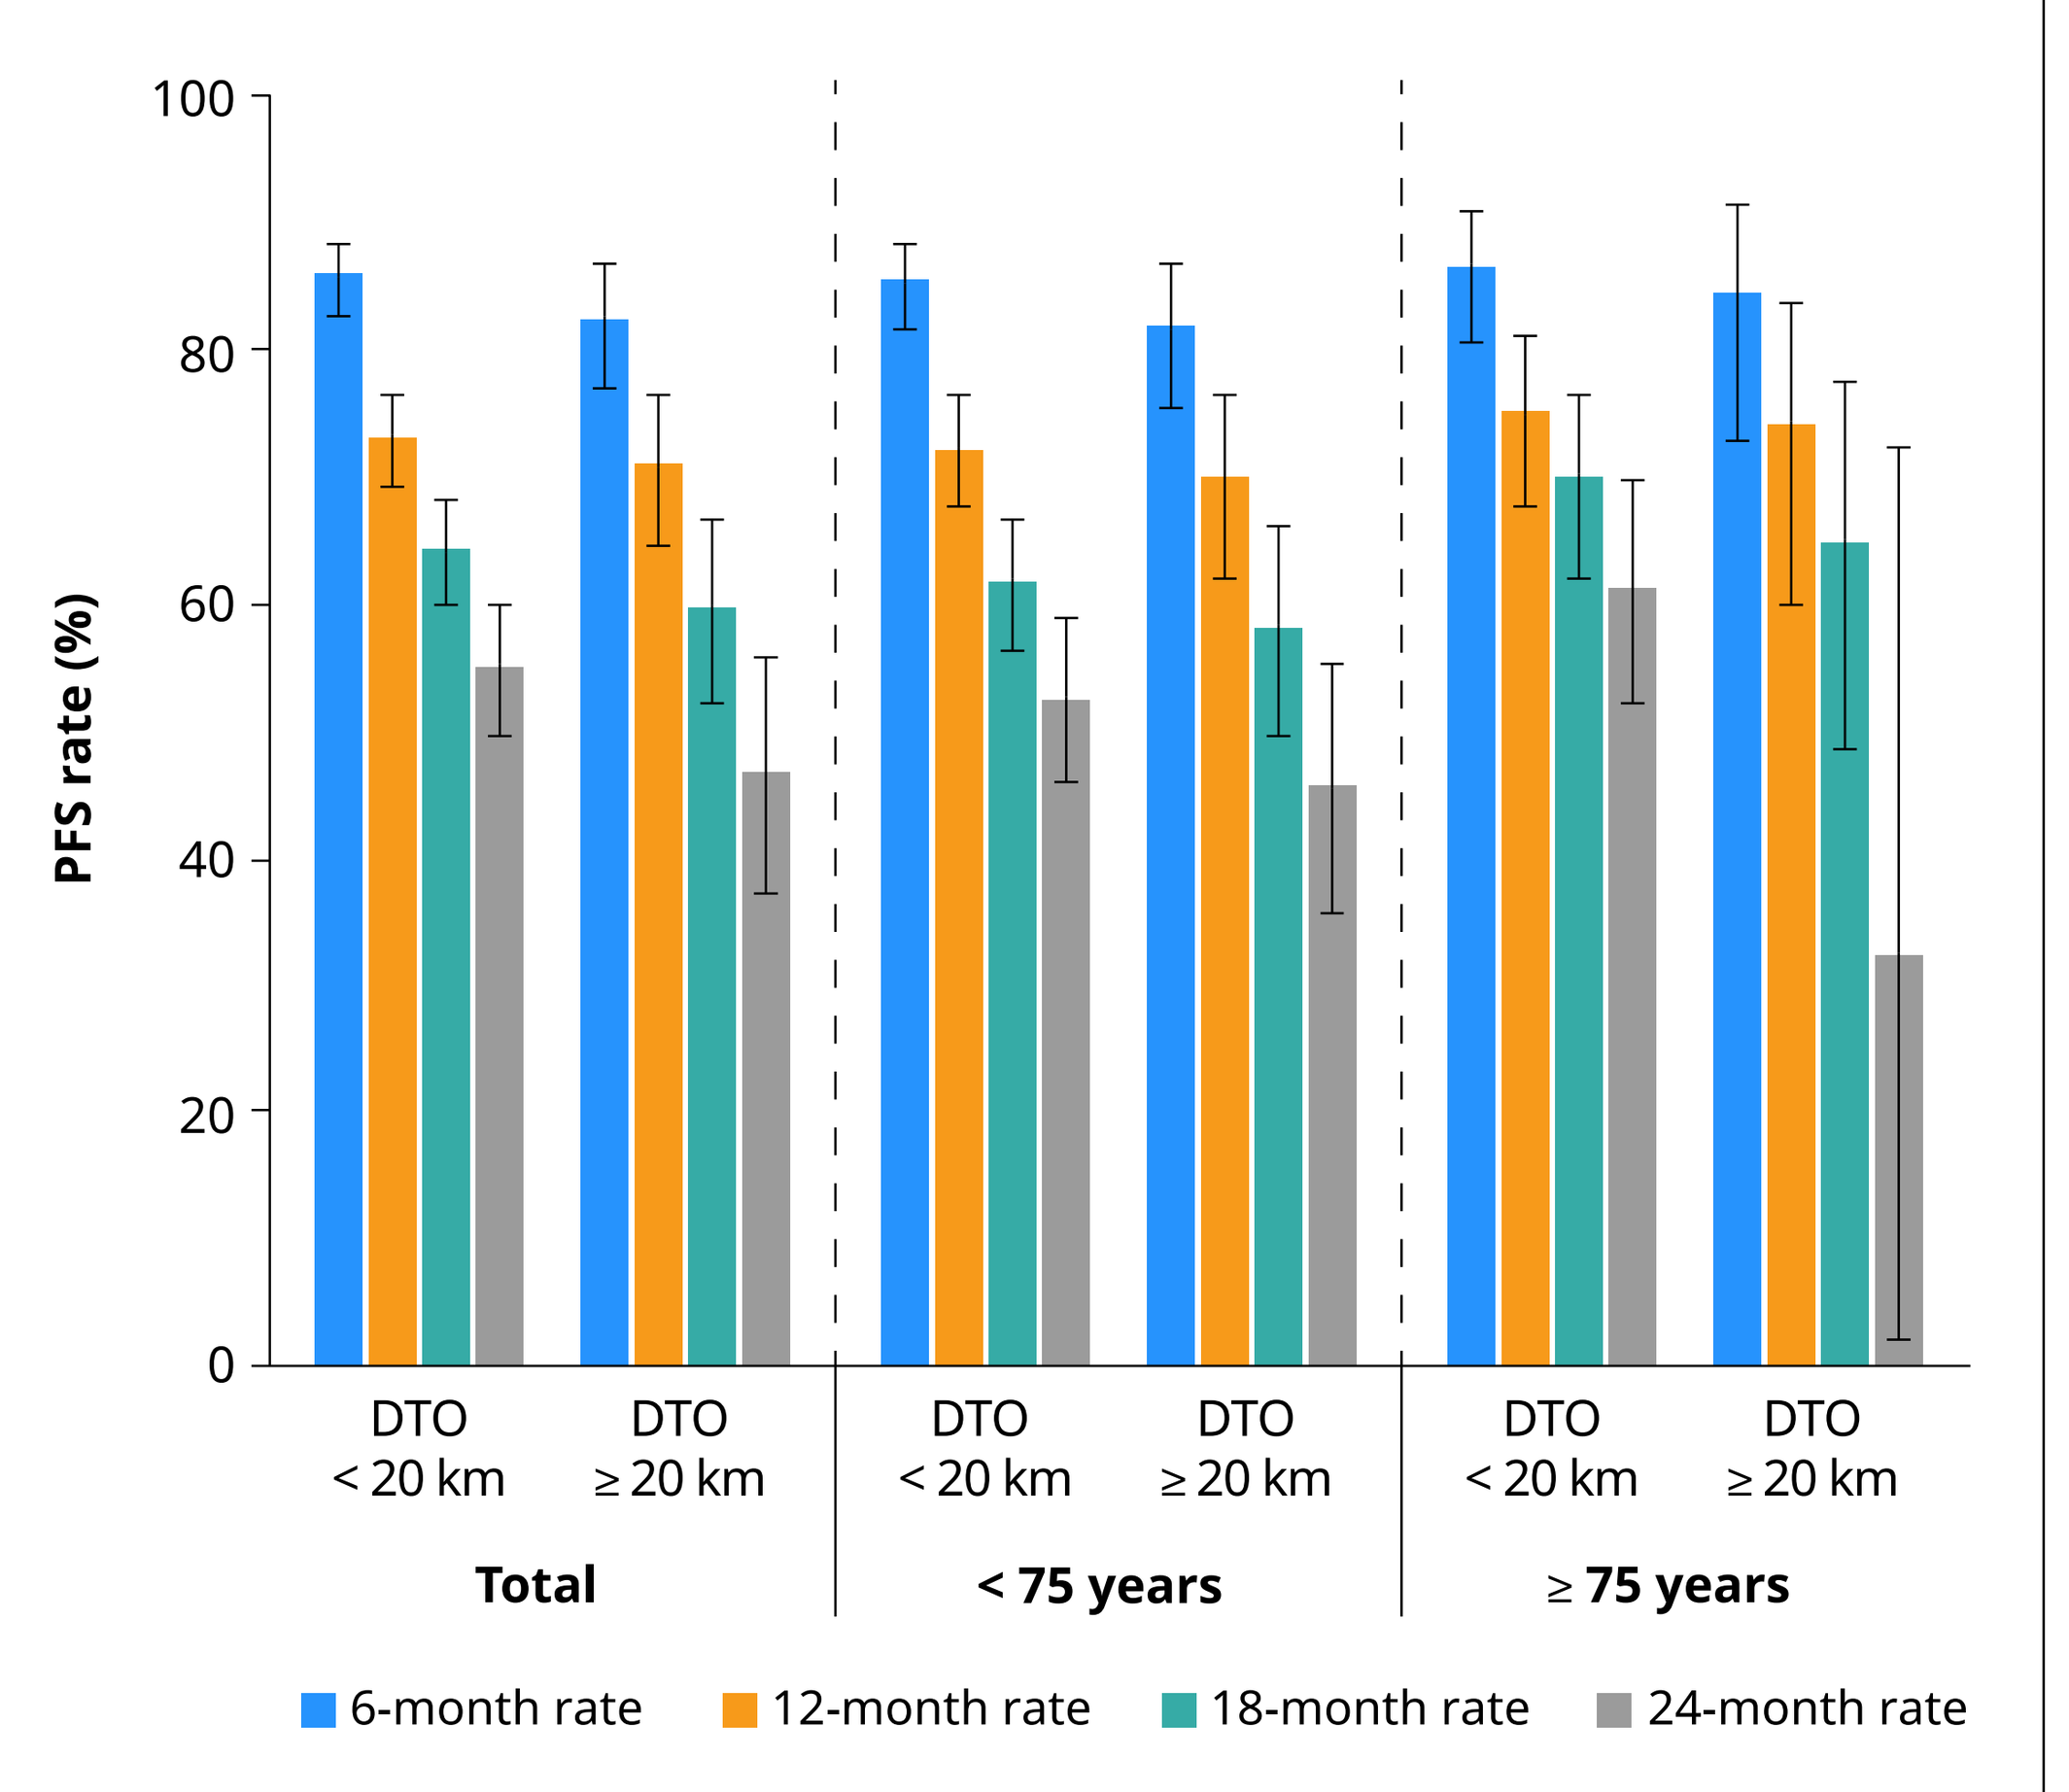
PFS rate (%)
100
80
60
40
20
0
DTO < 20 km
DTO ≥ 20 km
Total
< 75 years
≥ 75 years
6-month rate
12-month rate
18-month rate
24-month rate

DTO, distance to treating oncologist; PFS, progression-free survival.

LIMITATIONS

- Results and conclusions are not necessarily transferable to other countries and healthcare systems.
- Socioeconomic/demographic factors, such as DTO, are documented only at one timepoint (inclusion).
- Generally, further follow-up is needed.
- When examining small subgroups of patients without controlling for other variables, it is crucial to acknowledge that factors such as age, a higher percentage of visceral metastases or recurrent disease, and other potentially confounding variables may significantly influence outcome and safety.

Therefore, all provided analyses must be regarded as purely descriptive and exploratory, as they do not allow causal conclusions. Hypotheses derived from these results warrant further confirmatory investigation.

DISCUSSION

A larger proportion of PERFORM patients live within 20 km from their treating oncologist at start of 1L treatment. Patients of this subgroup show a tendency to be older, and they appear to be more likely to live alone compared to the smaller subgroup with a DTO of ≥ 20 km. Interestingly, especially elderly patients in the latter subgroup tend to present more frequently with *de novo* ABC. This might potentially be affected by a more rural residence and increasing travel burden of those patients – an effect that has been described before.[11] Patients under 75 years of age with a DTO < 20 km had a slightly higher observed frequency of visceral metastases. In line with other study results, NIS PERFORM has previously reported that non-de novo disease and visceral metastasis appear to be associated with a poorer prognosis, poorer response, and a tendency to progress earlier with 1L endocrine-based therapy.[8,12] Although these characteristics tend to be more common in the subgroup with shorter DTO, PFS-rates are comparable or even slightly higher in these patients compared to those with longer DTO, regardless of age.

Presenting author: Michael P. Lux

Email for more information: m.lux@vincenz.de

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster.

References: **1.** Finn RS, et al. *N Eng J Med*. 2016;375(20);1925–1936. **2.** Rugo HS, et al. *Breast Cancer Res Treat*. 2019;174(3):719–729. **3.** Turner, N. C. et al. *N Eng J Med*. 2018;379(20):1926–1936. **4.** Cristofanilli M, et al. *Lancet Oncol*. 2016;17(4),425–439. **5.** Slamon DJ, et al. *J Clin Oncol*. 2024;42(9):994–1000. **6.** Ruckhäberle E, et al. *Geburtshilfe und Frauenheilkunde*. 2024;84(9):813–836. **7.** Lux MP, et al. *Future Oncol*. 2022;18(36):3971-3982. **8.** Korrell, M. et al. *Ann Oncol*. 2024;9 (suppl_4):1-47. Abstract 209P. **9.** Bjelic-Radisic V, et al. 2024. Poster P045 presented at the 43rd Annual conference of the German Society of Senology (DGS) (Dresden). **10.** Pfeiler G, et al. *Ann Oncol*. 2024;35:S366. Abstract 356P. **11.** de Almeida RJ, et al. *Scientific reports*. 2022;12(1):8357. **12.** Brufsky A, et al. *Cancers*. 2023;15(21):5268.

Acknowledgments: The non-interventional study PERFORM is sponsored by Pfizer Pharma GmbH. Editorial support was provided by Dominique J. Verlaan, PhD, of Oxford PharmaGenesis Inc., Newtown, PA, USA, and was funded by Pfizer Inc.

Presented at the San Antonio Breast Cancer Symposium® • December 10–13, 2024 • San Antonio, TX

This presentation is the intellectual property of the author/presenter. Contact Michael P. Lux for permission to reprint and/or distribute.
Copyright © 2024. All rights reserved.

EM-DEU-plb-0146